Abstract 1823
(F-2062)

A Multi-center Global Surveillance Study of the *In Vitro* Activity of Retapamulin (SB-275833), A Novel Topical Pleuromutilin Against 2950 Clinical Isolates of *S. aureus* and Coagulase Negative Staphylococci from Uncomplicated Skin and Skin Structure Infections (SSSIs)

T. Stevens,[1] B. Johnson,[1] S. Bouchillon,[1] D. Hoban,[1] N. Scangarella,[2] R. Shawar,[2] J. Johnson,[1] R. Badal[1]

[1]International Health Management Associates, Schaumburg, IL, USA; [2]GlaxoSmithKline, Collegeville, PA, USA

Correspondence:
T. Stevens
IHMA
2122 Palmer Drive
Schaumburg
IL 60173
USA
Tel: +1 847 303 5003
Fax: +1 847 303 5601
E-mail: tstevens@ihmainc.com

Revised Abstract

Background: Retapamulin is a novel pleuromutilin currently in development as a topical antimicrobial for the treatment of skin and skin structure infections (SSSIs). Retapamulin has a unique mode of action, shows no target specific cross-resistance to other classes of antibiotics and is fully active against skin bacterial isolates carrying resistance determinants to established agents including β-lactams, macrolides, quinolones, fusidic acid and mupirocin. **Methods**: Clinical isolates of *Staphylococcus aureus* n = 1975, and coagulase negative staphylococci (CoNS), including *Staphylococcus epidermidis* n = 975, were collected from 53 sites in 13 countries during 2004 and 2005. All isolates were sent to the central laboratory for identification confirmation and testing. Susceptibility testing was performed using broth microdilution panels. All testing and quality controls (each day of testing) were conducted following Clinical and Laboratory Standards Institute (CLSI, formerly the National Committee for Clinical Laboratory Standards [NCCLS]) guidelines. **Results:** All antimicrobials were tested against *S. aureus*/CoNS isolates resulting in MIC_{90}s (expressed as µg/mL) of 0.12/0.06 for retapamulin; 4/>256 for mupirocin; 4/32 for gentamicin; 64/16 for neomycin; 2/8 for fusidic acid; >128/>128 for bacitracin; >32/>32 for clindamycin; >32/>32 for erythromycin; 32/32 for tetracycline; 4/2 for linezolid; 16/4 for cephalothin; 64/32 for ceftriaxone; 32/16 for penicillin; 4/16 for cloxacillin; and 16/8 for amoxicillin/clavulanic acid, respectively. **Conclusions:** Retapamulin had the lowest MIC_{90} values of all agents tested against all staphylococcal isolates and demonstrated greater activity against *S. aureus* (including methicillin-susceptible and -resistant strains) and CoNS isolates compared with topical and oral antimicrobial agents commonly used in the treatment of uncomplicated SSSIs.

Introduction

Retapamulin (SB-275833; Figure 1), a novel, semi-synthetic derivative of the pleuromutilins, a new class of antimicrobials, is currently in development for the topical treatment of a variety of Gram positive pathogens associated with secondarily-infected traumatic lesions and secondarily-infected dermatoses. The pleuromutilins are potent inhibitors of protein synthesis in bacteria through the interference of peptide bond formation by binding to the peptidyl transferase center of the 50S ribosomal subunit.[1] Due to the unique pleuromutilin mode of action, retapamulin shows no target specific cross-resistance to other classes of antibacterials.

Pleuromutilin derivatives have demonstrated significant *in vitro* activity against a variety of fastidious and non-fastidious bacterial pathogens, especially methicillin-resistant *Staphylococcus aureus* and coagulase-negative staphylococci.[2] Retapamulin was studied in a multi-center global surveillance program to determine the *in vitro* activity of this compound against a large diverse population of clinical isolates of *S. aureus* and coagulase-negative staphylococci from skin and skin structure infections (SSSIs).

Materials and Methods

- MIC endpoints were determined by broth microdilution according to Clinical and Laboratory Standards Institute (CLSI, formerly the National Committee for Clinical Laboratory Standards [NCCLS]) guidelines[3] for retapamulin and 14 comparators in customized dried broth microdilution panels (Trek Diagnostic Systems Ltd, West Sussex, UK). Comparator antimicrobial agents included: amoxicillin-clavulanic acid, cephalothin, clindamycin, fusidic acid, linezolid, mupirocin and tetracycline.
- Mueller-Hinton broth (Sensititre®, Cleveland, OH, USA) was used for all *Staphylococcus* species.
- The trays were incubated at 35°C in ambient air for 16–20 h before reading the MIC endpoints.
- Quality control testing was performed each day of testing as specified by the CLSI using the following isolates: *S. aureus* ATCC 29213 and *S. aureus* ATCC 25923. In addition, quality control ranges previously determined for retapamulin were used as a control.[4]
- All study organisms were clinical hospital- and community-acquired isolates collected and frozen at -70°C from March 2004 to March 2005 from 53 sites in 13 countries: Australia, Belgium, Canada, France, Germany, India, Italy, Mexico, Singapore, South Africa, Spain, the UK and the USA. All isolates were obtained from SSSIs from both adult and paediatric patients, with one isolate per patient. Organism collection, transport, confirmation of organism identification, antimicrobial susceptibility testing, as well as construction and management of a centralized database, were co-ordinated by International Health Management Associates, Inc. (IHMA, Schaumburg, IL, USA).
- The total number of isolates, MIC_{50} (µg/mL), MIC_{90} (µg/mL) and MIC ranges were determined for all antimicrobial agents tested. Resistant phenotypes and interpretive criteria were defined by the susceptibility of the corresponding antimicrobial agent according to CLSI breakpoints or the literature (mupirocin and fusidic acid only). Methicillin-resistance was based upon oxacillin screening agar.

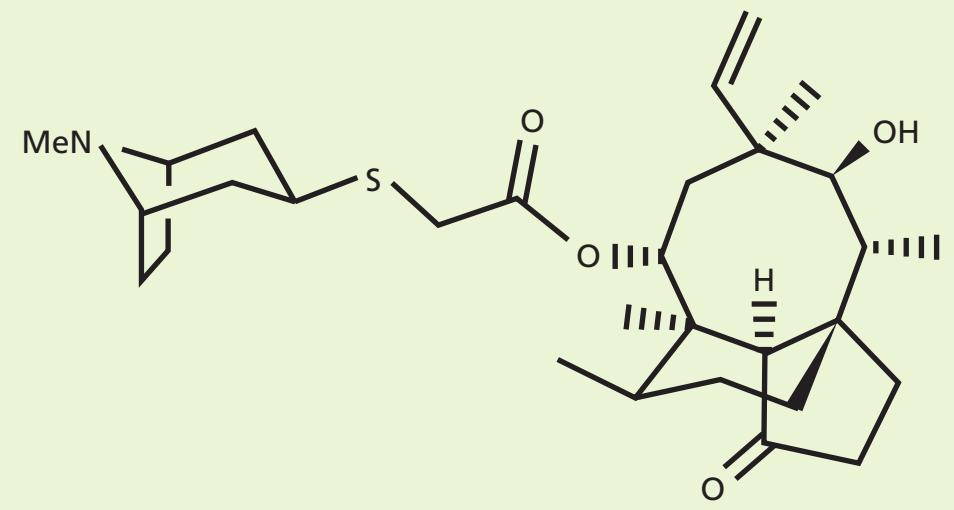

MeN
S
O
O
OH
H
O

Figure 1. Chemical Structure of Retapamulin

Results

The results are shown in Tables 1–3.

Table 1. MIC (µg/mL) Summary for Retapamulin and Comparators Against *S. aureus* (n = 1975)

				MIC (µg/mL)		
Compound[a]	%Sus	%Int	%Res	Range	MIC_{50}	MIC_{90}
Retapamulin	NA	NA	NA	0.004–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[b]	64.4	0	35.6	≤0.015–>32	1	16
Cephalothin	66.0	0.3	33.7	0.06–>256	0.5	16
Clindamycin	75.2	3.2	21.5	0.03–>32	0.12	>32
Fusidic acid	89.4	3.8	6.8	0.03–>32	0.12	2
Linezolid	100	0	0	0.5–4	2	4
Mupirocin	90.2	0	9.8	0.06–>256	0.25	4
Tetracycline	79.7	3.1	17.2	≤0.06–>32	0.5	32

[a]Interpretive criteria of compounds defined in the CLSI document M100-S15 where available;[5] Mupirocin susceptibility defined in Finlay *et al*.[6] Fusidic acid susceptibility defined in Toma and Barriault;[7] Note: β-lactams reported as resistant for MRSA in accordance with current CLSI guidelines.
[b]Amoxicillin/clavulanic acid was tested in a 2:1 ratio; MICs are reported based on the amoxicillin concentration.
Sus, susceptible; Int, intermediate; Res, resistant; NA, not available.

Table 2. MIC (µg/mL) Summary for Retapamulin and Comparators Against *S. aureus* Categorized by Various Antimicrobial Phenotypes.

				MIC (µg/mL)		
Compound[a]	%Sus	%Int	%Res	Range	MIC_{50}	MIC_{90}
Methicillin-susceptible[b] (n = 1326)						
Retapamulin	NA	NA	NA	0.015–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	95.9	0	4.1	≤0.015–>32	0.5	2
Cephalothin	98.3	0.5	1.2	0.06–64	0.5	1
Clindamycin	88.0	2.6	9.4	0.03–>32	0.12	2
Fusidic acid	89.4	3.9	6.6	0.06–>32	0.12	2
Linezolid	100	0	0	0.5–4	2	4
Mupirocin	92.8	0	7.2	0.06–>256	0.25	1
Tetracycline	83.9	2.9	13.3	≤0.06–>32	0.5	16
Methicillin-resistant[b] (n = 649)						
Retapamulin	NA	NA	NA	0.004–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	0	0	100	0.12–>32	8	32
Cephalothin	0	0	100	0.25–>256	4	64
Clindamycin	49.2	4.5	46.4	0.06–>32	1	>32
Fusidic acid	89.4	3.5	7.1	0.03–>32	0.12	2
Linezolid	100	0	0	0.5–4	2	2
Mupirocin	84.7	0	15.3	0.12–>256	0.25	16
Tetracycline	71.3	3.5	25.1	0.12–>32	0.5	>32
Macrolide-susceptible[d] (n = 835)						
Retapamulin	NA	NA	NA	0.004–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	92.3	0	7.7	≤0.015–32	0.5	2
Cephalothin	93.2	0.2	6.6	0.06–32	0.5	1
Clindamycin	96.8	2.0	1.2	0.03–>32	0.12	0.25
Fusidic acid	90.9	3.5	5.6	0.06–>32	0.12	1
Linezolid	100	0	0	0.5–4	2	4
Mupirocin	96.8	0	3.2	0.06–>256	0.25	0.5
Tetracycline	88.7	2.0	9.2	≤0.06–>32	0.5	8
Macrolide-intermediate[d] (n = 184)						
Retapamulin	NA	NA	NA	0.03–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	76.6	0	23.4	0.06–32	1	8
Cephalothin	78.8	0	21.2	0.12–128	0.5	8
Clindamycin	84.2	8.7	7.1	0.06–>32	0.12	1
Fusidic acid	84.2	6.0	9.8	0.06–32	0.25	2
Linezolid	100	0	0	1–4	2	4
Mupirocin	89.1	0	10.9	0.12–>256	0.25	8
Tetracycline	75.0	6.5	18.5	0.12–>32	0.5	32
Macrolide-resistant[d] (n = 956)						
Retapamulin	NA	NA	NA	0.015–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	37.6	0	62.4	0.12–>32	4	16
Cephalothin	39.9	0.4	59.7	0.12–>256	2	64
Clindamycin	54.7	3.2	42.1	0.06–>32	0.25	>32
Fusidic acid	89.1	3.7	7.2	0.03–>32	0.12	2
Linezolid	100	0	0	0.5–4	2	4
Mupirocin	84.6	0	15.4	0.06–>256	0.25	16
Tetracycline	72.8	3.3	23.8	0.12–>32	0.5	32
Mupirocin-susceptible[e] (n = 1781)						
Retapamulin	NA	NA	NA	0.004–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	66.7	0	33.3	≤0.015–>32	1	16
Cephalothin	68.4	0.3	31.3	0.06–256	0.5	16
Clindamycin	79.1	2.9	18.0	0.03–>32	0.12	>32
Fusidic acid	89.7	3.4	6.9	0.03–>32	0.12	2
Linezolid	100	0	0	0.5–4	2	4
Mupirocin	100	0	0	0.06–4	0.25	0.5
Tetracycline	81.5	2.8	15.7	≤0.06–>32	0.5	32
Mupirocin-resistant[e] (n = 194)						
Retapamulin	NA	NA	NA	0.03–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	42.8	0	57.2	0.06–>32	4	32
Cephalothin	44.3	0.5	55.2	0.25–>256	2	128
Clindamycin	39.7	6.7	53.6	0.06–>32	8	>32
Fusidic acid	86.6	7.2	6.2	0.06–>32	0.25	2
Linezolid	100	0	0	0.5–4	2	2
Mupirocin	0	0	100	8–>256	64	>256
Tetracycline	63.9	5.7	30.4	0.25–>32	1	>32
Fusidic acid-susceptible[f] (n = 1766)						
Retapamulin	NA	NA	NA	0.015–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	64.6	0	35.4	≤0.015–>32	1	16
Cephalothin	66.4	0.2	33.4	0.06–>256	0.5	16
Clindamycin	76.7	2.9	20.4	0.03–>32	0.12	>32
Fusidic acid	100	0	0	0.03–1	0.12	0.5
Linezolid	100	0	0	0.5–4	2	4
Mupirocin	90.5	0	9.5	0.06–>256	0.25	4
Tetracycline	81.0	2.8	16.2	≤0.06–>32	0.5	32
Fusidic acid-resistant[f] (n = 134)						
Retapamulin	NA	NA	NA	0.004–0.5	0.06	0.12
Amoxicillin-clavulanic acid[c]	59.7	0	40.3	0.12–>32	1	16
Cephalothin	63.4	1.5	35.1	0.12–>256	0.5	32
Clindamycin	63.4	5.2	31.3	0.06–>32	0.12	>32
Fusidic acid	0	0	100	4–>32	4	32
Linezolid	100	0	0	0.5–4	2	4
Mupirocin	91.0	0	9.0	0.12–>256	0.25	4
Tetracycline	63.4	7.5	29.1	0.12–>32	0.5	32

[a]Interpretive criteria of compounds defined in the CLSI document M100-S15, where available;[5]
Mupirocin susceptibility defined in Finlay *et al*.;[6] Fusidic acid susceptibility defined in Toma and Barriault;[7]
Note: β-lactams reported as resistant for MRSA in accordance with current CLSI guidelines.
[b]Phenotype is determined by the susceptibility of *S. aureus* to oxacillin as defined in CLSI document M100-S15 (2005).[5]
[c]Amoxicillin/clavulanic acid was tested in a 2:1 ratio; MICs are reported based on the amoxicillin concentration.
[d]Phenotype is determined by the susceptibility of *S. aureus* to erythromycin as defined in CLSI document M100-S15 (2005).[5]
[e]Phenotype is determined by the susceptibility of *S. aureus* to mupirocin (≤4 susceptible, ≥8 resistant) as defined in Finlay *et al*.[6]
[f]Phenotype is determined by the susceptibility of *S. aureus* to fusidic acid (≤1 susceptible, ≥4 resistant) as defined in Toma and Barriault.[7]
Sus, susceptible; Int, intermediate; Res, resistant; NA, not available.

Table 3. MIC (µg/mL) Summary for Retapamulin and Comparators Against Coagulase-negative Staphylococci (n = 975)

				MIC (µg/mL)		
Compound[a]	%Sus	%Int	%Res	Range	MIC_{50}	MIC_{90}
Retapamulin	NA	NA	NA	≤0.002–0.5	0.06	0.06
Amoxicillin-Clavulanic acid[b]	39.5	0	60.5	≤0.015–>32	1	8
Cephalothin	40.4	0	59.6	≤0.03–256	0.5	4
Clindamycin	61.1	2.4	36.5	≤0.015–>32	0.12	>32
Fusidic acid	73.9	3.4	22.7	≤0.015–>32	0.12	8
Linezolid	100	0	0	0.03–4	1	2
Mupirocin	65.0	0	35.0	0.06–>256	0.25	>256
Tetracycline	73.0	4.4	22.6	≤0.06–>32	1	32
Methicillin-susceptible[b] (n = 397)						
Retapamulin	NA	NA	NA	≤0.002–0.5	0.06	0.06
Amoxicillin-Clavulanic acid[c]	97.0	0	3.0	≤0.015–16	0.25	2
Cephalothin	99.2	0	0.8	≤0.03–64	0.25	1
Clindamycin	79.1	3.0	17.9	≤0.015–>32	0.06	>32
Fusidic acid	86.6	0.8	12.6	≤0.015–32	0.12	8
Linezolid	100	0	0	0.03–4	1	2
Mupirocin	83.1	0	16.9	0.06–>256	0.25	64
Tetracycline	82.9	3.5	13.6	≤0.06–>32	0.5	16
Methicillin-resistant[b] (n = 578)						
Retapamulin	NA	NA	NA	0.015–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	0	0	100	0.12–>32	2	8
Cephalothin	0	0	100	0.12–256	1	8
Clindamycin	48.8	1.9	49.3	≤0.015–>32	2	>32
Fusidic acid	65.2	5.2	29.6	≤0.015–>32	0.12	16
Linezolid	100	0	0	0.25–4	1	2
Mupirocin	52.6	0	47.4	0.06–>256	1	>256
Tetracycline	66.3	5.0	28.7	0.12–>32	2	>32
Mupirocin-susceptible[d] (n = 634)						
Retapamulin	NA	NA	NA	≤≤0.002–0.5	0.06	0.06
Amoxicillin-Clavulanic acid[c]	51.1	0	48.9	≤0.015–>32	0.5	4
Cephalothin	51.7	0	48.3	≤0.03–256	0.5	2
Clindamycin	77.3	2.2	20.5	≤0.015–>32	0.12	>32
Fusidic acid	78.4	2.8	18.8	0.03–>32	0.12	8
Linezolid	100	0	0	0.25–4	1	2
Mupirocin	100	0	0	0.06–4	0.12	0.5
Tetracycline	80.8	2.4	16.9	≤0.06–>32	1	32
Mupirocin-resistant[d] (n = 341)						
Retapamulin	NA	NA	NA	0.015–0.5	0.06	0.12
Amoxicillin-Clavulanic acid[c]	17.9	0	82.1	0.06–>32	2	16
Cephalothin	19.4	0	80.6	0.12–128	1	16
Clindamycin	31.1	2.6	66.3	≤0.015–>32	32	>32
Fusidic acid	65.7	4.4	29.9	≤0.015–>32	0.12	16
Linezolid	100	0	0	0.03–4	1	2
Mupirocin	0	0	100	8–>256	>256	>256
Tetracycline	58.7	8.2	33.1	0.12–>32	2	>32

[a]Interpretive criteria of compounds defined in the CLSI document M100-S15, where available;[5]
Mupirocin susceptibility defined in Finlay *et al*.;[6] Fusidic acid susceptibility defined in Toma and Barriault;[7]
Note: β-lactams reported as resistant for methicillin-resistant staphylococci in accordance with current CLSI guidelines.
[b]Phenotype is determined by the susceptibility of *S. aureus* to oxacillin as defined in CLSI document M100-S15 (2005).[5]
[c]Amoxicillin/clavulanic acid was tested in a 2:1 ratio; MICs are reported based on the amoxicillin concentration.
[d]Phenotype is determined by the susceptibility of *S. aureus* to mupirocin (≤4 susceptible, ≥8 resistant) as defined in Finlay *et al*.[6]
Sus, susceptible; Int, intermediate; Res, resistant; NA, not available.

Conclusions

- Retapamulin was 16–32-fold more active than all comparators tested with a MIC_{90} of ≤0.12 µg/mL with 0.5 µg/mL inhibiting all 2950 staphylococcal strains tested.
- The global rate of mupirocin and fusidic acid-resistance in *S. aureus* isolates from SSSI infections was 10% and 7%, respectively. For coagulase-negative staphylococci, 35% were mupirocin-resistant and 23% were fusidic acid-resistant.
- Retapamulin was the most active compound tested in this study with an MIC_{90} of 0.12 µg/mL for *S. aureus* and 0.06 µg/mL for coagulase-negative staphylococci, which was 16- to 32-fold lower than that of the nearest comparator.
- Retapamulin retained its *in vitro* activity *versus S. aureus* resistant to methicillin, erythromycin, fusidic acid, or mupirocin, with MIC_{90}s of 0.12 µg/mL for all strains, regardless of resistant phenotype.
- Retapamulin retained its *in vitro* activity *versus* coagulase-negative staphylococci isolates resistant to either methicillin or mupirocin, with MIC_{90}s ≤0.12 µg/mL regardless of the resistant phenotype.
- The *in vitro* activity of retapamulin *versus S. aureus* and coagulase-negative staphylococci isolated from SSSI could potentially make retapamulin a useful addition to the therapeutic options for the treatment of SSSIs.
- These *in vitro* findings strongly support retapamulin's clinical potential, and further trial data are required in order to assess its efficacy in patients.

References

1 Schlunzen F, Pyetan E, Fucini P *et al*. *Mol Microbiol* 2004; 54: 1287–1294.
2 Jones RN, Pfaller MA, Rhomberg PR, Walter DH. *J Clin Microbiol* 2002; 40: 461–465.
3 CLSI. Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standard, 6th Edition. Document M7-A6. Wayne, PA, USA: CLSI, 2005.
4 Jones RN. SB-275833, An Investigational Topical Agent: Quality Control Studies for the MIC (M7-A6) Method. Document on file, UH2004/00009/00, GlaxoSmithKline, Collegeville, PA, USA, 2005.
5 CLSI. Performance Standards for Antimicrobial Susceptibility Testing, in Document M100-S15. Wayne, PA, USA: CLSI, 2005.
6 Finlay JE, Miller LA, Poupard JA. *Antimicrob Agents Chemother* 1997. 41: 1137–1139.
7 Toma E, Barriault D. *J Clin Microbiol* 1995; 33: 1712–1715.

Acknowledgements

This study was sponsored by a grant from GlaxoSmithKline.