FRIDAY – AAR-543

Global *In vitro* Surveillance of Eravacycline Against Gram-negative and Gram-positive Clinical Isolates, Including Multidrug-Resistant Pathogens, Collected in 2017

Sara Hwang[1], Ekaterina Efimova[1], Corey Fyfe[1], Stephen Hawser[2], Ian Morrissey[2]

[1]Tetraphase Pharmaceuticals, Watertown, MA, [2]IHMA Europe Sàrl, Monthey, Switzerland

Contact:
Tetraphase Pharmaceuticals Medical Information
tetraphase@druginfo.com
(833) 793-7282

Introduction

Eravacycline is a novel, fully-synthetic, fluorocycline antibacterial of the tetracycline class approved by the FDA and EMA for the treatment of complicated intra-abdominal infections (cIAIs) in patients ≥18 years of age. Eravacycline is active against Gram-negative pathogens and many Gram-positive and anaerobic bacteria found in cIAI, and retains activity against the most common tetracycline-specific acquired resistance mechanisms (i.e., efflux and ribosomal protection)[1]. The purpose of this study was to evaluate the *in vitro* activity of eravacycline and comparators against Enterobacteriaceae, *Acinetobacter baumannii*, *Stenotrophomonas maltophilia, Staphylococcus aureus* (including methicillin-resistant *S. aureus*, MRSA) and *Enterococcus* spp., including those that are multidrug-resistant (MDR).

Methods

- A total of 8755 clinical isolates collected from hospitals globally in 2017 from genitourinary, gastrointestinal, body fluid, and respiratory sources were tested.
- MDR was defined as resistance to ≥3 from cefepime/cefotaxime/ceftazidime/ceftriaxone (any one), aztreonam, gentamicin, a carbapenem (meropenem or ertapenem), levofloxacin, piperacillin-tazobactam, tetracycline or tigecycline, according to CLSI/FDA or EUCAST breakpoints.
- Minimal inhibitory concentration (MIC) values were determined by broth microdilution according to the CLSI guidelines for eravacycline and comparators.
- Quality control testing was performed each day of testing as specified by the CLSI.
- Antibiotic susceptibility was determined using CLSI 2019 breakpoints[2], except for eravacycline and tigecycline where FDA breakpoints[3] were used, and EUCAST 2019 breakpoints[4].

Results

Figure 1. Percent Distribution of all Isolates by Geographic Origin

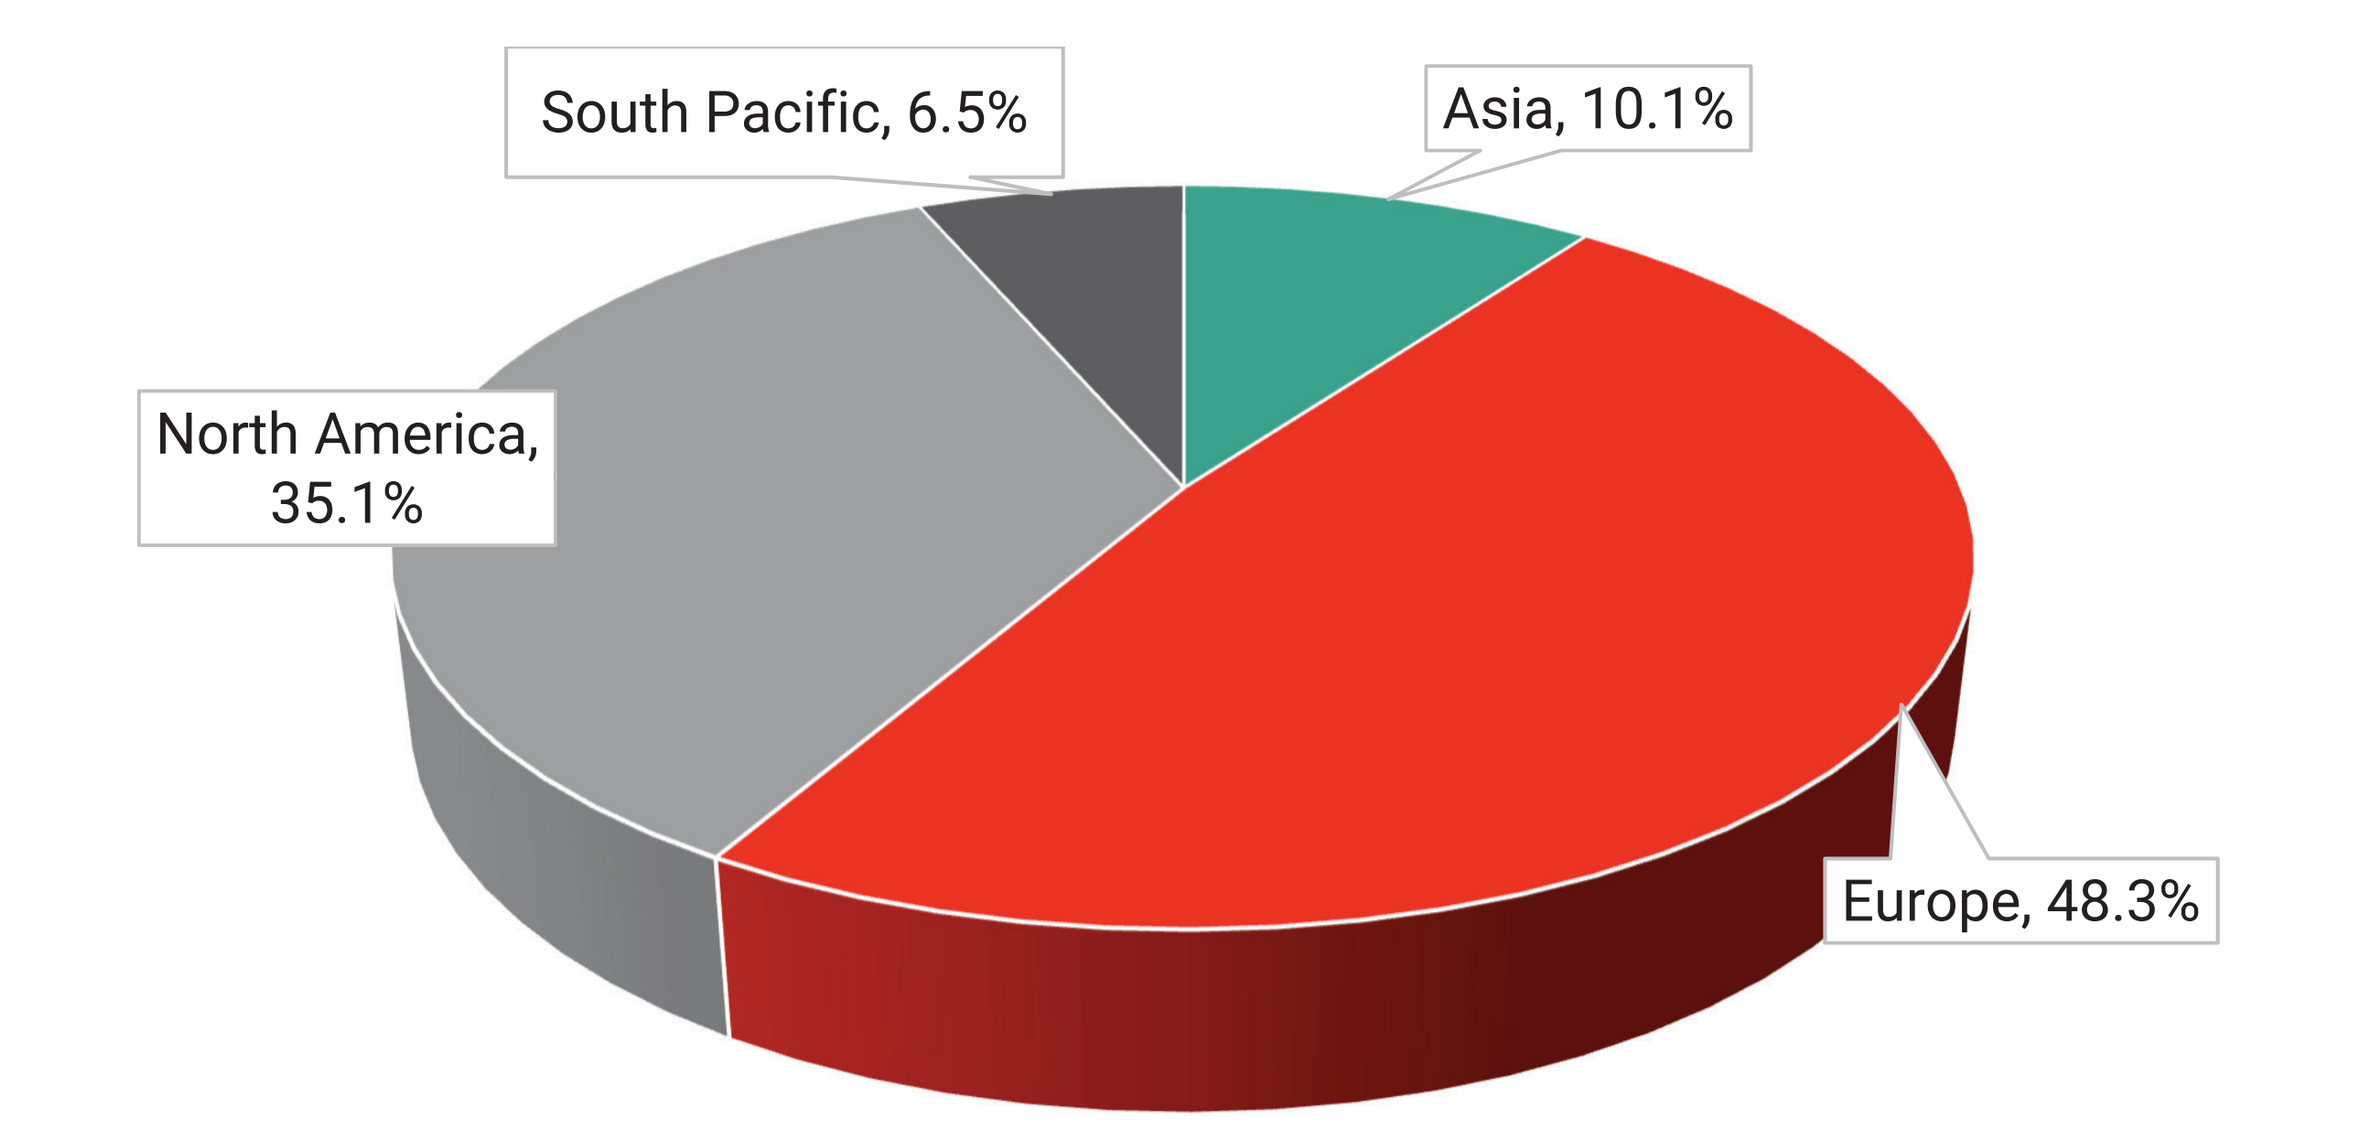

South Pacific, 6.5%
Asia, 10.1%
North America, 35.1%
Europe, 48.3%

Results (cont'd)

- Global distribution of the clinical isolates is shown in Figure 1.
- Susceptibility data, $MIC_{50/90}$ values, and MIC ranges for eravacycline and comparators are shown in Tables 1 – 6.
- Eravacycline $MIC_{50/90}$ values for all Enterobacteriaceae were 0.25/0.5 μg/mL, and for MDR Enterobacteriaceae were 0.25/2 μg/mL [Table 1].
- The overall susceptibility of eravacycline for Enterobacteriaceae and MDR Enterobacteriaceae was high, and did not differ much between FDA and EUCAST breakpoint interpretations, which were 93% and 85%, respectively [Table 1].
- Clinically important Gram-negative isolates, such as *C. freundii, E. cloacae, E. coli* and *K. pneumoniae*, were highly susceptible to eravacycline, demonstrating susceptibilities of 92.33%, 90.59%, 99.15% and 85.74%, respectively [Table 2].
- Eravacycline had high susceptibility rates, >90%, for Gram-positive organisms [Tables 3 and 4]. For MRSA, eravacycline susceptibility rates were 86.30% and 97.95% when using the FDA and EUCAST breakpoint interpretations, respectively [Table 3].
- Eravacycline MIC_{90} values for *S. maltophilia* [Table 5] and *A. baumannii*, including carbapenem-resistant *A. baumannii* (CRAB) [Table 6], were 1 μg/mL.
- For all organisms tested, eravacycline MIC_{90} values were primarily 2- to 8-fold lower than tigecycline values.

Table 1. Susceptibility of Enterobacteriaceae, Including MDR Isolates, to Eravacycline and Comparators

Organism	Drug	$MIC_{50/90}$	Range	FDA/CLSI Breakpoints (S\|I\|R)	%S	%R	EUCAST Breakpoints (S\|I\|R)	%S	%R
Enterobacteriaceae[&] (n=2074) MDR (n=329[1],539[2], 536[3])	**Eravacycline**	0.25/0.5 0.25/2	0.03 - 16 0.06 - 8	≤0.5 \| -- \| --*	92.86% 84.79%	-- --	≤0.5 \| -- \| ≥1	92.86% 84.70%	7.14% 15.30%
	Amikacin	1/4 2/8	≤0.25 - >64 ≤0.25 - >64	≤16 \| 32 \| ≥64	98.94% 96.10%	0.77% 2.97%	≤8 \| 16 \| ≥32	97.69% 91.42%	1.06% 3.92%
	Aztreonam	0.12/>16 >16/>16	≤0.03 - >16 ≤0.03 - >16	≤4 \| 8 \| ≥16	74.49% 16.70%	23.43% 79.22%	≤1 \| 2-4 \| ≥8	71.79% 6.53%	25.51% 90.67%
	Cefepime	0.06/>16 16/>16	≤0.008 - >16 0.015 - >16	≤2 \| 4-8 \| ≥16	81.63% 32.28%	14.03% 52.69%	≤1 \| 2-4 \| ≥8	78.74% 21.83%	15.86% 58.77%
	Ceftazidime	0.25/128 64/>128	≤0.03 - >128 0.06 - >128	≤4 \| 8 \| ≥16	76.18% 24.12%	21.65% 69.94%	≤1 \| 2-4 \| ≥8	73.00% 9.33%	23.82% 83.02%
	Ceftriaxone	0.12/>4 >4/>4	≤0.015 - >4 ≤0.015 - >4	≤1 \| 2 \| ≥4	72.28% 13.36%	27.15% 85.90%	≤1 \| 2 \| ≥4	72.28% 5.78%	27.15% 93.66%
	Ertapenem	0.008/0.5 0.12/>2	≤0.002 - >2 0.004 - >2	≤0.5 \| 1 \| ≥2	92.19% 72.91%	5.11% 18.92%	≤0.5 \| -- \| ≥1	92.19% 70.71%	7.81% 29.29%
	Gentamicin	0.25/16 1/>16	≤0.12 - >16 ≤0.12 - >16	≤4 \| 8 \| ≥16	87.56% 55.84%	11.81% 42.49%	≤2 \| 4 \| ≥8	86.93% 56.16%	12.44% 41.79%
	Levofloxacin	0.06/>8 8/>8	0.008 - >8 0.008 - >8	≤0.5 \| 1 \| ≥2	77.77% 32.47%	19.29% 61.22%	≤0.5 \| 1 \| ≥2	77.77% 36.94%	19.29% 57.46%
	Meropenem	0.03/0.12 0.06/4	0.008 - >4 0.015 - >4	≤1 \| 2 \| ≥4	96.48% 86.64%	3.18% 12.06%	≤2 \| 4-8 \| ≥16	96.82% 87.69%	-- --
	Minocycline	2/16 4/>16	≤0.12 - >16 0.25 - >16	≤4 \| 8 \| ≥16	82.26% 58.63%	10.13% 23.56%	No Breakpoints Defined	-- --	-- --
	Piperacillin-tazobactam	2/128 64/>128	≤0.25 - >128 ≤0.25 - >128	≤16/4 \| 32/4-64/4 \| ≥128/4	82.21% 44.34%	11.57% 41.74%	≤8 \| 16 \| ≥32	78.64% 31.16%	17.79% 63.06%
	Tetracycline	1/>64 64/>64	≤0.25 - >64 ≤0.25 - >64	≤4 \| 8 \| ≥16	76.52% 39.52%	21.36% 56.40%	No Breakpoints Defined	-- --	-- --
	Tigecycline	0.5/2 1/4	0.03 - 32 0.12 - 8	≤2 \| 4 \| ≥8*	95.81% 92.95%	1.06% 2.41%	≤0.5 \| -- \| ≥1	69.43% 51.31%	30.57% 48.69%

[&]*Citrobacter freundii, Enterobacter cloacae, Escherichia coli, Klebsiella oxytoca*, and *Klebsiella pneumoniae* only; [1]FDA; [2]CLSI; [3]EUCAST; *FDA breakpoint; $MIC_{50/90}$ – minimum inhibitory concentration required to inhibit growth of 50/90% of isolates; %S, %R, percent susceptible or resistant; n – number of isolates; MDR – multi-drug resistant; Units in μg/mL

Table 2. Susceptibility of *Citrobacter freundii, Escherichia coli, Klebsiella penumoniae* and *Enterobacter cloacae*, Including MDR Isolates, to Eravacycline and Comparators

Organism	Drug	$MIC_{50/90}$	Range	FDA/CLSI Breakpoints (S\|I\|R)	%S	%R	EUCAST Breakpoints (S\|I\|R)	%S	%R
C. freundii (n=287) MDR (n=44[1], 73[2], 85[3])	**Eravacycline**	0.25/0.5 0.25/2	0.06 - 2 0.12 - 2	≤0.5 \| -- \| --*	92.33% 77.27%	-- --	≤0.5 \| -- \| ≥1	92.33% 85.53%	7.67% 16.47%
	Ceftriaxone	0.25/>4 >4/>4	0.03 - >4 0.12 - >4	≤1 \| 2 \| ≥4	67.60% 12.33%	31.71% 86.30%	≤1 \| 2 \| ≥4	67.60% 3.53%	31.71% 95.29%
	Ertapenem	0.015/0.25 0.25/2	≤0.002 - >2 0.004 - >2	≤0.5 \| 1 \| ≥2	94.77% 80.82%	3.83% 15.07%	≤0.5 \| -- \| ≥1	94.77% 82.35%	5.23% 17.65%
	Levofloxacin	0.06/2 2/>8	0.015 - >8 0.03 - >8	≤0.5 \| 1 \| ≥2	82.58% 41.10%	14.98% 52.05%	≤0.5 \| 1 \| ≥2	82.58% 52.94%	14.98% 42.35%
	Meropenem	0.03/0.12 0.06/1	0.008 - >4 0.015 - >4	≤1 \| 2 \| ≥4	97.56% 90.41%	2.09% 8.22%	≤2 \| 4-8 \| ≥16	97.91% 92.94%	-- --
	Piperacillin-tazobactam	2/128 64/>128	1 - >128 2 - >128	≤16/4 \| 32/4-64/4 \| ≥128/4	77.00% 32.88%	11.50% 45.21%	≤8 \| 16 \| ≥32	72.82% 15.29%	23.00% 77.65%
	Tetracycline	1/>64 8/>64	≤0.25 - >64 ≤0.25 - >64	≤4 \| 8 \| ≥16	83.62% 49.32%	14.98% 49.32%	No Breakpoints Defined	-- --	-- --
	Tigecycline	0.5/2 0.5/2	0.12 - 4 0.25 - 4	≤2 \| 4 \| ≥8*	95.82% 90.91%	-- --	≤0.5 \| -- \| ≥1	69.69% 49.41%	30.31% 50.59%
E. cloacae (n=404) MDR (n=84[1], 120[2], 141[3])	**Eravacycline**	0.25/0.5 0.5/2	0.12 - 8 0.25 - 8	≤0.5 \| -- \| --*	90.59% 77.38%	-- --	≤0.5 \| -- \| ≥1	90.59% 82.98%	9.41% 17.02%
	Ceftriaxone	0.5/>4 >4/>4	≤0.015 - >4 0.5 - >4	≤1 \| 2 \| ≥4	57.43% 0.83%	42.08% 99.17%	≤1 \| 2 \| ≥4	57.43% 0.71%	42.08% 99.29%
	Ertapenem	0.06/1 1/>2	0.008 - >2 0.03 - >2	≤0.5 \| 1 \| ≥2	81.19% 46.67%	9.65% 30.83%	≤0.5 \| -- \| ≥1	81.19% 46.81%	18.81% 53.19%
	Levofloxacin	0.03/2 0.5/>8	0.008 - >8 0.015 - >8	≤0.5 \| 1 \| ≥2	86.39% 59.17%	10.64% 33.33%	≤0.5 \| 1 \| ≥2	86.39% 65.25%	10.64% 28.37%
	Meropenem	0.06/0.12 0.12/2	0.008 - >4 0.015 - >4	≤1 \| 2 \| ≥4	96.53% 89.17%	2.97% 9.17%	≤2 \| 4-8 \| ≥16	97.03% 91.49%	-- --
	Piperacillin-tazobactam	2/>128 128/>128	≤0.25 - >128 1 - >128	≤16/4 \| 32/4-64/4 \| ≥128/4	70.79% 20.00%	16.83% 56.67%	≤8 \| 16 \| ≥32	65.10% 9.93%	29.21% 82.98%
	Tetracycline	2/64 4/>64	0.5 - >64 1 - >64	≤4 \| 8 \| ≥16	82.18% 53.33%	14.85% 40.83%	No Breakpoints Defined	-- --	-- --
	Tigecycline	0.5/2 1/4	0.25 - 8 0.25 - 8	≤2 \| 4 \| ≥8*	93.56% 83.33%	1.49% 7.14%	≤0.5 \| -- \| ≥1	54.95% 47.52%	45.05% 52.58%
E. coli (n=471) MDR (n=76[1], 152[2], 120[3])	**Eravacycline**	0.12/0.25 0.25/0.25	0.03 - 1 0.06 - 1	≤0.5 \| -- \| --*	99.15% 98.68%	-- --	≤0.5 \| -- \| ≥1	99.15% 99.17%	0.85% 0.83%
	Ceftriaxone	0.06/>4 >4/>4	≤0.015 - >4 ≤0.015 - >4	≤1 \| 2 \| ≥4	74.52% 28.95%	25.27% 71.05%	≤1 \| 2 \| ≥4	74.52% 11.67%	25.27% 88.33%
	Ertapenem	0.008/0.03 0.015/0.25	≤0.002 - >2 0.004 - >2	≤0.5 \| 1 \| ≥2	98.30% 94.74%	0.42% 1.32%	≤0.5 \| -- \| ≥1	98.30% 93.33%	1.70% 6.67%
	Levofloxacin	0.12/>8 >8/>8	0.008 - >8 0.008 - >8	≤0.5 \| 1 \| ≥2	59.02% 13.16%	38.64% 85.53%	≤0.5 \| 1 \| ≥2	59.02% 5.83%	38.64% 93.33%
	Meropenem	0.015/0.03 0.03/0.06	0.008 - 2 0.015 - 2	≤1 \| 2 \| ≥4	99.79% 99.34%	-- --	≤2 \| 4-8 \| ≥16	100% 100%	-- --
	Piperacillin-tazobactam	2/8 2/64	≤0.25 - >128 ≤0.25 - >128	≤16/4 \| 32/4-64/4 \| ≥128/4	92.78% 83.55%	3.18% 7.24%	≤8 \| 16 \| ≥32	90.02% 75.00%	7.22% 21.67%
	Tetracycline	2/>64 >64/>64	≤0.25 - >64 0.5 - >64	≤4 \| 8 \| ≥16	61.15% 21.05%	37.58% 77.63%	No Breakpoints Defined	-- --	-- --
	Tigecycline	0.25/1 0.25/1	0.03 - 4 0.12 - 4	≤2 \| 4 \| ≥8*	99.15% 98.68%	-- --	≤0.5 \| -- \| ≥1	89.60% 88.33%	10.40% 11.67%
K. pneumoniae (n=484) MDR (n=104[1], 140[2], 134[3])	**Eravacycline**	0.25/1 0.5/2	0.06 - 8 0.12 - 8	≤0.5 \| -- \| --*	85.74% 69.23%	-- --	≤0.5 \| -- \| ≥1	85.74% 73.88%	14.26% 26.12%
	Ceftriaxone	0.06/>4 >4/>4	≤0.015 - >4 0.03 - >4	≤1 \| 2 \| ≥4	72.31% 9.29%	26.86% 88.57%	≤1 \| 2 \| ≥4	72.31% 5.97%	26.86% 92.54%
	Ertapenem	0.008/1 0.25/>2	0.004 - >2 0.008 - >2	≤0.5 \| 1 \| ≥2	89.05% 62.14%	9.50% 32.86%	≤0.5 \| -- \| ≥1	89.05% 61.19%	10.95% 38.81%
	Levofloxacin	0.06/>8 >8/>8	0.015 - >8 0.03 - >8	≤0.5 \| 1 \| ≥2	71.28% 14.29%	23.35% 74.29%	≤0.5 \| 1 \| ≥2	71.28% 13.43%	23.35% 76.12%
	Meropenem	0.03/0.12 0.06/>4	0.008 - >4 0.015 - >4	≤1 \| 2 \| ≥4	90.91% 68.57%	8.47% 29.29%	≤2 \| 4-8 \| ≥16	91.53% 69.40%	-- --
	Piperacillin-tazobactam	2/>128 128/>128	≤0.25 - >128 ≤0.25 - >128	≤16/4 \| 32/4-64/4 \| ≥128/4	80.58% 36.43%	15.08% 52.14%	≤8 \| 16 \| ≥32	76.45% 29.85%	19.42% 64.93%
	Tetracycline	2/>64 >64/>64	0.5 - >64 0.5 - >64	≤4 \| 8 \| ≥16	70.87% 37.14%	26.03% 57.14%	No Breakpoints Defined	-- --	-- --
	Tigecycline	1/2 1/4	0.25 – 16 0.25 - 8	≤2 \| 4 \| ≥8*	92.56% 89.42%	3.10% 5.77%	≤0.5 \| -- \| ≥1	48.55% 21.64%	51.45% 78.36%

[1]FDA; [2]CLSI; [3]EUCAST; *FDA breakpoint; $MIC_{50/90}$ – minimum inhibitory concentration required to inhibit growth of 50/90% of isolates; %S, %R, percent susceptible or resistant; n – number of isolates; MDR – multi-drug resistant; Units in μg/mL

Table 3. Susceptibility of *Staphylococcus aureus*, Including MRSA, to Eravacycline and Comparators

Organism	Drug	$MIC_{50/90}$	Range	FDA/CLSI Breakpoints (S\|I\|R)	%S	%R	EUCAST Breakpoints (S\|I\|R)	%S	%R
S. aureus (n=564)	**Eravacycline**	0.03/0.06	≤0.008 - 0.5	≤0.06 \| -- \| --*	90.96%	--	≤0.25 \| -- \| ≥0.5	98.94%	1.06%
	Clindamycin	0.12/>2	≤0.03 - >2	≤0.5 \| 1-2 \| ≥4	76.24%	23.40%	≤0.25 \| 0.5 \| ≥1	76.24%	23.76%
	Daptomycin	0.5/0.5	≤0.06 - 1	≤1 \| -- \| --	100%	--	≤1 \| -- \| ≥2	100%	--
	Levofloxacin	0.25/>4	0.06 - >4	≤1 \| 2 \| ≥4	58.33%	40.25%	≤1 \| -- \| ≥2	58.33%	41.67%
	Linezolid	1/2	≤0.5 - 2	≤4 \| -- \| ≥8	100%	--	≤4 \| -- \| ≥8	100%	--
	Oxacillin	>2/>2	≤0.06 - >2	≤2 \| -- \| ≥4	48.23%	51.77%	≤2 \| -- \| ≥4	48.23%	51.77%
	Tetracycline	0.25/>16	≤0.06 - >16	≤4 \| 8 \| ≥16	83.69%	15.78%	≤1 \| 2 \| ≥4	82.45%	16.67%
	Tigecycline	0.12/0.25	0.03 - 1	≤0.5 \| -- \| --*	99.29%	--	≤0.5 \| -- \| ≥1	99.29%	0.71%
	Vancomycin	0.5/1	≤0.25 - 2	≤2 \| 4-8 \| ≥16	100%	--	≤2 \| -- \| ≥4	100%	--
MSSA (n=272)	**Eravacycline**	0.03/0.06	≤0.008 - 0.25	≤0.06 \| -- \| --*	95.96%	--	≤0.25 \| -- \| ≥0.5	100%	--
	Clindamycin	0.12/0.12	≤0.03 - >2	≤0.5 \| 1-2 \| ≥4	93.75%	5.88%	≤0.25 \| 0.5 \| ≥1	93.75%	6.25%
	Daptomycin	0.5/0.5	0.12 - 1	≤1 \| -- \| --	100%	--	≤1 \| -- \| ≥2	100%	--
	Levofloxacin	0.12/0.5	0.06 - >4	≤1 \| 2 \| ≥4	90.81%	8.82%	≤1 \| -- \| ≥2	90.81%	9.19%
	Linezolid	1/2	≤0.5 - 2	≤4 \| -- \| ≥8	100%	--	≤4 \| -- \| ≥8	100%	--
	Oxacillin	0.25/0.5	≤0.06 – 2	≤2 \| -- \| ≥4	100%	--	≤2 \| -- \| ≥4	100%	--
	Tetracycline	0.25/0.5	≤0.06 - >16	≤4 \| 8 \| ≥16	93.01%	6.62%	≤1 \| 2 \| ≥4	92.28%	7.72%
	Tigecycline	0.12/0.25	0.03 - 1	≤0.5 \| -- \| --*	99.63%	--	≤0.5 \| -- \| ≥1	99.63%	0.37%
	Vancomycin	0.5/1	≤0.25 - 2	≤2 \| 4-8 \| ≥16	100%	--	≤2 \| -- \| ≥4	100%	--
MRSA (n=292)	**Eravacycline**	0.03/0.12	≤0.008 - 0.5	≤0.06 \| -- \| --*	86.30%	--	≤0.25 \| -- \| ≥0.5	97.95%	2.05%
	Clindamycin	0.12/>2	≤0.03 - >2	≤0.5 \| 1-2 \| ≥4	59.93%	39.73%	≤0.25 \| 0.5 \| ≥1	59.93%	40.07%
	Daptomycin	0.5/0.5	≤0.06 - 1	≤1 \| -- \| --	100%	--	≤1 \| -- \| ≥2	100%	--
	Levofloxacin	>4/>4	0.06 - >4	≤1 \| 2 \| ≥4	28.08%	69.52%	≤1 \| -- \| ≥2	28.08%	71.92%
	Linezolid	1/2	≤0.5 - 2	≤4 \| -- \| ≥8	100%	--	≤4 \| -- \| ≥8	100%	--
	Oxacillin	>2/>2	>2 - >2	≤2 \| -- \| ≥4	--	100%	≤2 \| -- \| ≥4	--	100%
	Tetracycline	0.25/>16	≤0.06 - >16	≤4 \| 8 \| ≥16	75.00%	24.32%	≤1 \| 2 \| ≥4	73.29%	25.00%
	Tigecycline	0.12/0.25	0.03 - 1	≤0.5 \| -- \| --*	98.97%	--	≤0.5 \| -- \| ≥1	98.97%	1.03%
	Vancomycin	0.5/1	≤0.25 - 2	≤2 \| 4-8 \| ≥16	100%	--	≤2 \| -- \| ≥4	100%	--

*FDA breakpoint; $MIC_{50/90}$ – minimum inhibitory concentration required to inhibit growth of 50/90% of isolates; %S, %R, percent susceptible or resistant; n – number of isolates; Units in μg/mL

Table 4. Susceptibility of *Enterococcus* spp., Including Vancomycin-Resistant *Enterococcus* (VRE), to Eravacycline and Comparators

Organism	Drug	$MIC_{50/90}$	Range	FDA/CLSI Breakpoints (S\|I\|R)	%S	%R	EUCAST Breakpoints (S\|I\|R)	%S	%R
Enterococcus spp. (n=778) VRE (n=173[1,2], 179[3])	**Eravacycline**	0.06/0.06 0.03/0.06	0.008 - 0.5 0.008 - 0.25	≤0.06 \| -- \| --*	93.70% 96.53%	-- --	≤0.12 \| -- \| ≥0.25	98.97% 98.88%	1.03% 1.12%
	Ampicillin	2/>8 >8/>8	≤0.25 - >8 0.5 - >8	≤8 \| -- \| ≥16	58.10% 11.56%	41.90% 88.44%	≤4 \| 8 \| ≥16	57.97% 13.41%	41.90% 86.59%
	Daptomycin	2/4 2/4	≤0.03 - >8 0.12 - 8	≤1 \| 2-4 \| ≥8	43.96% 78.03%	0.77% 2.31%	No Breakpoints Defined	-- --	-- --
	Levofloxacin	>8/>8 >8/>8	0.25 - >8 2 - >8	≤2 \| 4 \| ≥8	41.13% 0.58%	57.07% 99.42%	≤4 \| -- \| ≥8	42.93% 1.12%	57.07% 98.88%
	Linezolid	2/2 1/2	0.5 - >4 0.5 - 4	≤2 \| 4 \| ≥8	98.59% 98.84%	0.26% --	≤4 \| -- \| ≥8	99.74% 100%	0.26% --
	Penicillin	4/>8 >8/>8	≤0.06 - >8 0.5 - >8	≤8 \| -- \| ≥16	57.33% 12.14%	42.67% 87.86%	No Breakpoints Defined	-- --	-- --
	Tigecycline	0.12/0.25 0.12/0.12	0.015 - 1 0.03 - 1	≤0.25 \| -- \| --*	97.04% 97.69%	-- --	≤0.25 \| -- \| ≥0.5	97.04% 97.77%	2.96% 2.23%
	Vancomycin	1/>16 >16/>16	0.5 - >16 >16 - >16	≤4 \| 8-16 \| ≥32	76.99% --	22.24% 100%	≤4 \| -- \| ≥8	76.99% --	23.01% 100%
E. faecalis (n=409)	**Eravacycline**	0.06/0.06	0.008 - 0.5	≤0.06 \| -- \| --*	92.18%	--	≤0.12 \| -- \| ≥0.25	99.51%	0.49%
	Ampicillin	1/2	0.5 - >8	≤8 \| -- \| ≥16	99.51%	0.49%	≤4 \| 8 \| ≥16	99.51%	0.49%
	Daptomycin	1/2	≤0.03 - 4	≤2 \| 4 \| ≥8	97.80%	--	No Breakpoints Defined	--	--
	Levofloxacin	1/>8	0.5 - >8	≤2 \| 4 \| ≥8	69.44%	30.32%	≤4 \| -- \| ≥8	69.68%	30.32%
	Linezolid	2/2	0.5 - >4	≤2 \| 4 \| ≥8	99.27%	0.49%	≤4 \| -- \| ≥8	99.51%	0.49%
	Penicillin	2/8	1 - >8	≤8 \| -- \| ≥16	98.29%	1.71%	No Breakpoints Defined	--	--
	Tigecycline	0.12/0.25	0.015 - 1	≤0.25 \| -- \| --*	97.80%	--	≤0.25 \| -- \| ≥0.5	97.80%	2.20%
	Vancomycin	1/2	0.5 - >16	≤4 \| 8-16 \| ≥32	94.62%	4.40%	≤4 \| -- \| ≥8	94.62%	5.38%
E. faecium (n=369)	**Eravacycline**	0.03/0.06	0.008 - 0.25	≤0.06 \| -- \| --*	95.39%	--	≤0.12 \| -- \| ≥0.25	98.37%	1.63%
	Ampicillin	>8/>8	≤0.25 - >8	≤8 \| -- \| ≥16	12.20%	87.80%	≤4 \| 8 \| ≥16	11.92%	87.80%
	Daptomycin	2/4	≤0.03 - >8	≤2 \| 4 \| ≥8	65.85%	1.63%	No Breakpoints Defined	--	--
	Levofloxacin	>8/>8	0.25 - >8	≤2 \| 4 \| ≥8	9.76%	86.72%	≤4 \| -- \| ≥8	13.28%	86.72%
	Linezolid	2/2	0.5 - 4	≤2 \| 4 \| ≥8	97.83%	--	≤4 \| -- \| ≥8	100%	--
	Penicillin	>8/>8	≤0.06 - >8	≤8 \| -- \| ≥16	11.92%	88.08%	No Breakpoints Defined	--	--
	Tigecycline	0.06/0.12	0.015 - 1	≤0.25 \| -- \| --*	96.21%	--	≤0.25 \| -- \| ≥0.5	96.21%	3.79%
	Vancomycin	1/>16	0.5 - >16	≤4 \| 8-16 \| ≥32	57.45%	42.01%	≤4 \| -- \| ≥8	57.45%	42.55%

[1]FDA; [2]CLSI; [3]EUCAST; *FDA breakpoint; $MIC_{50/90}$ – minimum inhibitory concentration required to inhibit growth of 50/90% of isolates; %S, %R, percent susceptible or resistant; n – number of isolates; Units in μg/mL

Table 5. Susceptibility of *Stenotrophomonas maltophilia* to Eravacycline and Comparators

Organism	Drug	$MIC_{50/90}$	Range	FDA/CLSI Breakpoints (S\|I\|R)	%S	%R	EUCAST Breakpoints (S\|I\|R)	%S	%R
S. maltophilia (n=51)	**Eravacycline**	0.5/1	0.06 - 1	No Breakpoints Defined	--	--	No Breakpoints Defined	--	--
	Amikacin	>64/>64	0.5 - >64	No Breakpoints Defined	--	--	No Breakpoints Defined	--	--
	Ceftazidime	16/>64	1 - >64	≤8 \| 16 \| ≥32	47.06%	35.29%	No Breakpoints Defined	--	--
	Levofloxacin	1/8	0.06 - 8	≤2 \| 4 \| ≥8	84.31%	11.76%	No Breakpoints Defined	--	--
	Meropenem	>64/>64	16 - >64	No Breakpoints Defined	--	--	No Breakpoints Defined	--	--
	Minocycline	0.25/1	0.06 - 2	≤4 \| 8 \| ≥16	100%	--	No Breakpoints Defined	--	--
	Piperacillin-tazobactam	128/>128	16 - >128	No Breakpoints Defined	--	--	No Breakpoints Defined	--	--
	Tetracycline	16/32	2 - >64	No Breakpoints Defined	--	--	No Breakpoints Defined	--	--
	Tigecycline	1/2	0.25 - 8	No Breakpoints Defined	--	--	No Breakpoints Defined	--	--
	Trimethoprim Sulfa	0.25/2	0.06 - 64	≤2/38 \| -- \| ≥4/76	92.16%	7.84%	≤4 \| -- \| ≥8	98.04%	1.96%

$MIC_{50/90}$ – minimum inhibitory concentration required to inhibit growth of 50/90% of isolates; %S, %R, percent susceptible or resistant; n – number of isolates; Units in μg/mL

Table 6. Susceptibility of *Acinetobacter baumannii*, Including Carbapenem-Resistant *Acinetobacter baumannii* (CRAB) Isolates, to Eravacycline and Comparators

Organism	Drug	$MIC_{50/90}$	Range	FDA/CLSI Breakpoints (S\|I\|R)	%S	%R	EUCAST Breakpoints (S\|I\|R)	%S	%R
A. baumannii (n=497) CRAB (286[2], 280[3])	**Eravacycline**	0.25/1 0.5/1	≤0.015 - 4 0.03 - 4	No Breakpoints Defined	-- --	-- --	No Breakpoints Defined	-- --	-- --
	Amikacin	64/>64 >64/>64	0.12 - >64 0.5 - >64	≤16 \| 32 \| ≥64	47.48% 16.43%	50.70% 80.42%	≤8 \| 16 \| ≥32	45.07% 13.21%	52.52% 85.00%
	Ampicillin-sulbactam	32/>64 64/>64	0.25 - >64 8 - >64	≤8/4 \| 16/8 \| ≥32/16	38.03% 2.45%	53.92% 87.41%	No Breakpoints Defined	-- --	-- --
	Cefepime	64/>64 >64/>64	≤0.03 - >64 8 - >64	≤8 \| 16 \| ≥32	29.58% 0.70%	64.99% 96.50%	No Breakpoints Defined	-- --	-- --
	Ceftazidime	>64/>64 >64/>64	0.5 - >64 4 - >64	≤8 \| 16 \| ≥32	33.80% 2.10%	63.78% 96.15%	No Breakpoints Defined	-- --	-- --
	Colistin	0.5/1 0.5/2	0.12 - >32 0.25 - >32	≤2 \| -- \| ≥4	93.96% 91.26%	6.04% 8.74%	≤2 \| -- \| ≥4	93.96% 91.07%	6.04% 8.93%
	Levofloxacin	16/64 32/>64	≤0.03 - >64 0.25 - >64	≤2 \| 4 \| ≥8	30.99% 0.35%	65.19% 96.15%	≤0.5 \| 1 \| ≥2	30.38% --	69.22% 100%
	Meropenem	32/>64 >64/>64	0.06 - >64 8 - >64	≤2 \| 4 \| ≥8	37.42% --	57.55% 100%	≤2 \| 4-8 \| ≥16	37.42% --	56.34% 100%
	Minocycline	1/16 4/16	≤0.03 - >64 ≤0.03 - >64	≤4 \| 8 \| ≥16	69.22% 53.15%	17.30% 26.92%	No Breakpoints Defined	-- --	-- --
	Tetracycline	16/>64 >64/>64	0.25 - >64 0.5 - >64	≤4 \| 8 \| ≥16	31.19% 6.64%	59.96% 86.01%	No Breakpoints Defined	-- --	-- --
	Tigecycline	2/8 4/8	0.06 - >16 0.25 - >16	No Breakpoints Defined	-- --	-- --	No Breakpoints Defined	-- --	-- --

[2]CLSI; [3]EUCAST; $MIC_{50/90}$ – minimum inhibitory concentration required to inhibit growth of 50/90% of isolates; %S, %R, percent susceptible or resistant; n – number of isolates; Units in μg/mL

Conclusions

- Eravacycline exhibited potent *in vitro* activity against Enterobacteriaceae, *A. baumannii* and clinically important Gram-positive organisms, including resistant pathogens collected globally in 2017.
- Eravacycline demonstrated 2- to 4-fold lower MIC_{90} values compared to tigecycline against Enterobacteriaceae, *S. aureus, Enterococcus* spp, and *S. maltophilia*, including resistant organisms.
- This 2017 global surveillance investigation highlights eravacycline's broad spectrum potency against Gram-negative and Gram-positive bacteria, including MDR strains, and further underscores its potential benefit as an empiric treatment of intra-abdominal infections caused by resistant pathogens.

References

1. XERAVA™ (eravacycline) [package insert]. Watertown, MA; Tetraphase Pharmaceuticals; 2018.
2. Clinical and Laboratory Standards Institute (CLSI). Performance Standards for Antimicrobial Susceptibility Testing. 29th ed. CLSI supplement M100. Wayne, PA: Clinical and Laboratory Standards Institute; 2019
3. U.S. Food and Drug Administration (FDA). Antibacterial Susceptibility Test Interpretive Criteria, 2018. http://www.fda.gov
4. The European Committee on Antimicrobial Susceptibility Testing, 2019 (EUCAST). Breakpoint tables for interpretation of MICs and zone diameters. Version 9.0 http://www.eucast.org

ASM Microbe, June 20 – June 24, 2019, San Francisco, CA